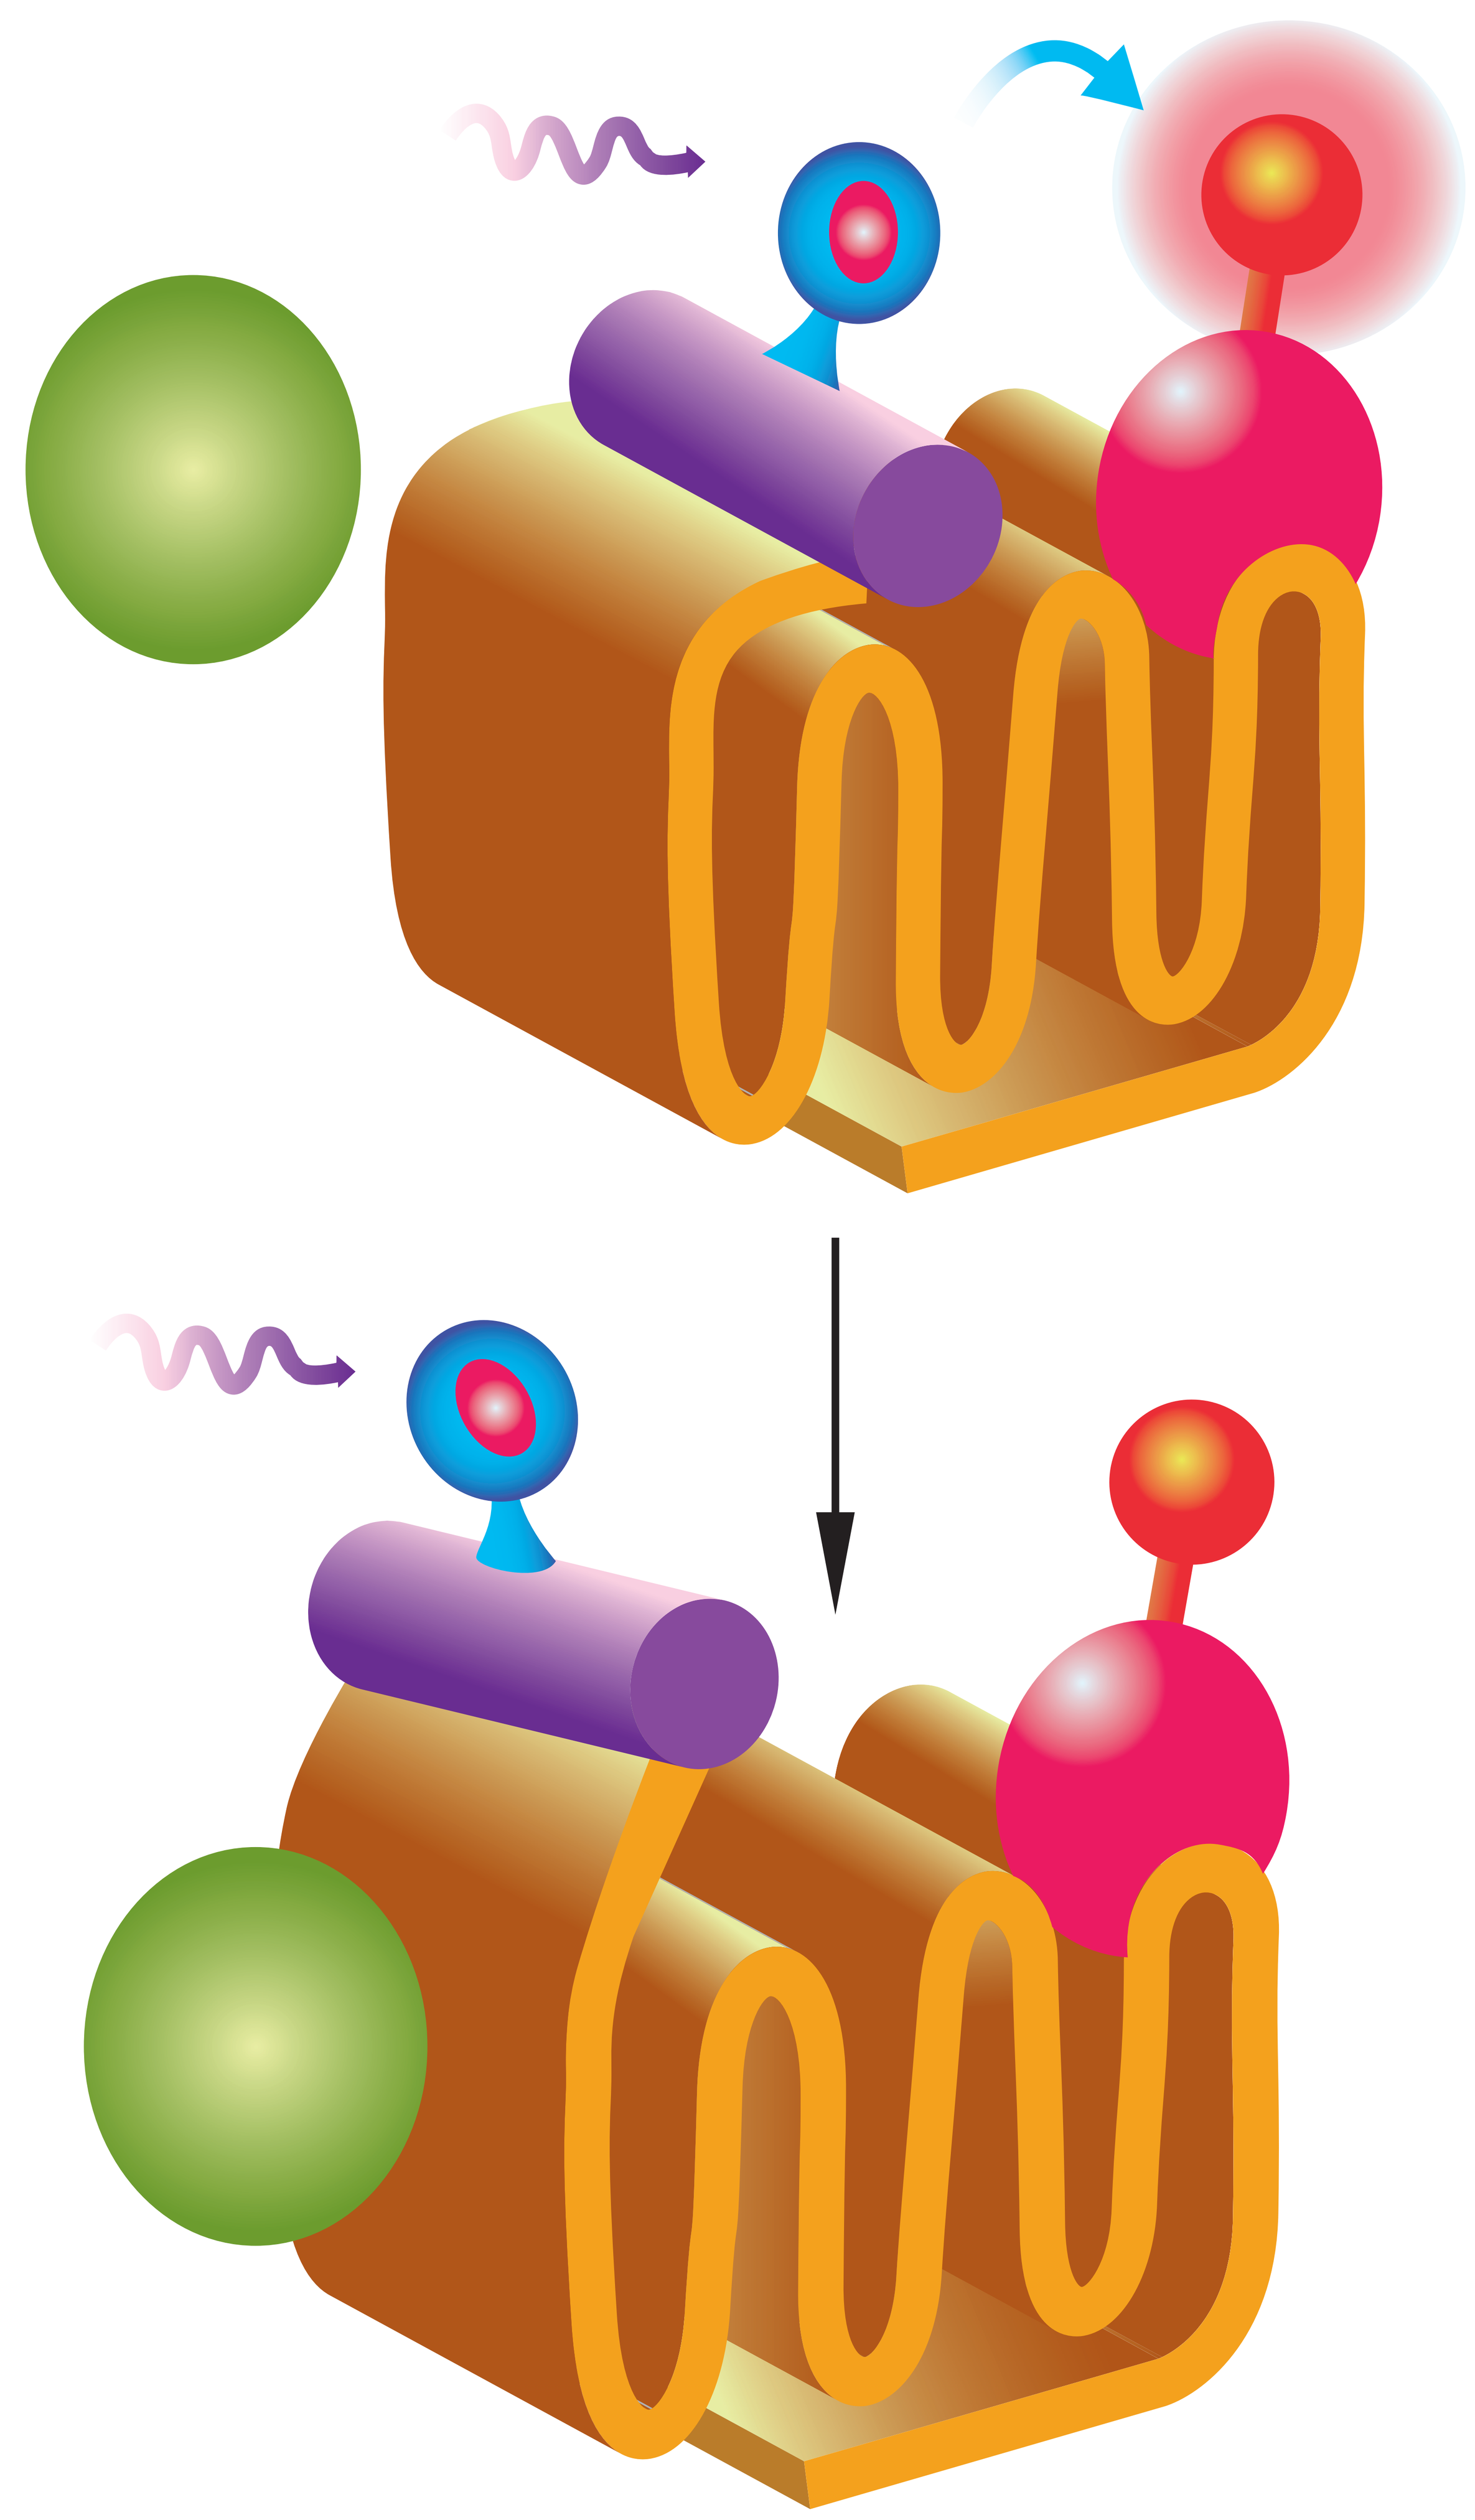

CANNABIDIOL ACTS AS AN ALLOSTERIC MODULATOR OF CANNABINOID CB_2 RECEPTORS

Eva Martínez-Pinilla[1], Katia Varani[2], Irene Reyes-Resina[3,4], Edgar Angelats[3,4], Salvatore Casano[5], Fabrizio Vincenzi[2], Carolina Sánchez-Carnerero Callado[5], Verónica Sánchez de Medina[5], Carlos Ferreiro-Vera[5], Enric I. Canela[3,4], José Luis Lanciego[6], Xavier Nadal[5], Gemma Navarro[3,4,7] Pier Andrea Borea[2], and Rafael Franco[3,4]

1. Departamento de Morfología y Biología Celular, Universidad de Oviedo, Oviedo, Asturias, Spain. 2. Department of Medical Sciences, University of Ferrara, Ferrara, Ferrara, Italy. 3. Department of Biochemistry and Molecular Biomedicine, Universitat de Barcelona, Barcelona, Catalunya, Spain. 4. Centro de Investigación en Red, Enfermedades Neurodegenerativas (CIBERNED), Instituto de Salud Carlos III, Madrid, Madrid, Spain. 5. Phytoplant Research S.L. Córdoba. Córdoba, Spain. 6. Neuroscience Department, University of Navarra, Pamplona, Navarra, Spain. 7. Department of Biochemistry and Physiology, Universitat de Barcelona, Barcelona, Catalunya, Spain.

ABSTRACT

In a heterologous expression system consisting of membrane preparations of cells (HEK-293) expressing the CB_2R human version, we have confirmed using a synthetic radiolabeled agonist, [3H]-WIN 55,212-2, that CBD does not bind with high affinity to the orthosteric site of human CB_2Rs. CBD was, however, able to negatively modulate in forskolin-induced intracellular cAMP determination assays the effect of JWH133, a selective CB_2R agonist. These results were consistent with CBD-mediated significant modification in the dissociation kinetics of the binding of a fluorophore-conjugated CB_2R-selective compound, detected using the HTRF (homogenous time-resolved fluorescence resonance energy transfer) methodology. In conclusion, some of the actions of CBD may be mediated by interaction with an allosteric site in CB2R that allows modulation of the action of receptor agonists.

MATERIALS AND METHODS

Radioligand binding assays

Competition binding experiments were performed in CHO cells expressing human CB_2Rs (CHO-CB_2R) by incubation with 3 nM [3H]-WIN 55,212-2 and different concentrations of the tested compounds with membranes obtained from CHO-CB_2R cells (10 µg protein per sample) for 60 minutes at 30°C. Non-specific binding was determined in the presence of 1 µM WIN 55,212-2. Bound and free radioactivity were separated by filtering the assay mixture through Whatman GF/B glass fibre filters using a Brandel cell harvester. The filter-bound radioactivity was counted using a Packard Tri Carb 2810 TR scintillation counter (Perkin Elmer).

Non-radioligand binding assays

For fluorescent ligand-binding assays, human embryonic kidney 293T (HEK-293T) were transfected with the SNAP-CB_2R plasmid and labelled with SNAP-Lumi4-Tb. In the competition-binding assay cells were incubated with 100 nM red CB_2R ligand (labelled CM-157), in the presence of increasing concentrations (0-10 µM range) of CBD. Saturation binding experiments were performed by incubating cells expressing Tb-labelled SNAP-CB_2R with increasing concentrations of the red CB_2R ligand (range, 0-300 nM final). For each concentration, non-specific binding was determined by adding 100 µM unlabelled CB_2R ligand. Signal was detected using an EnVision microplate reader (Perkin-Elmer, Waltham, MA, USA) equipped with a FRET optic module allowing donor excitation at 337 nm and signal collection at both 665 and 620 nm.

Kinetic binding assays were performed by incubating cells expressing Tb-labelled SNAP-CB_2R with 24 nM or 48 nM red CB_2R ligand. After short centrifugation, association was measured using the EnVision microplate reader and HTRF® signals were obtained in intervals of 60 seconds. After association, dissociation was induced by the quick addition of a large excess of ligands (unlabelled CM-157, CBD or both) to a final concentration of 5 µM. Once again, fluorescent signals corresponding to the competitive binding were recorded in intervals of 6 seconds.

cAMP determination

For cAMP determinations, serum-starved cells were detached and resuspended in growing medium containing 50 µM zardaverine and plated in 384-well microplates (2,500 cells/well), pretreated (15 minutes) with the corresponding antagonists -or vehicle- and stimulated with agonists (15 minutes) before adding 0.5 µM forskolin or vehicle. HTRF measures after 15 minutes of incubation were performed using the Lance Ultra cAMP kit (PerkinElmer, Waltham, MA, USA). Fluorescence at 665 nm was analysed on a PHERAstar Flagship microplate reader equipped with an HTRF optical module (BMG Lab technologies, Offenburg, Germany).

Statistical analysis

Data results from at least 3 independent experiments. The data in the graphs are the mean ± SEM. Statistical analysis was performed with SPSS 18.0 software. Significant differences were considered when $p<0.05$.

RESULTS

Figure 1. Competition curves in radioligand and HTRF-based assays
(A) Competition curves of specific [3H]-WIN 55,212-2 binding by unlabeled WIN 55,212-2, CBD and THC. Data is expressed as the mean ± SEM of three independent experiments performed in duplicate.
(B) Competition curve of specific binding of 100 nM red CB_2R ligand with increasing concentrations of CBD (0-10 µM) measured by HTRF. Data represents the mean ± SEM of a representative experiment performed in quadruplicate. HTRF ratio = 665 nm acceptor signal/620 nm donor signal x 10,000.

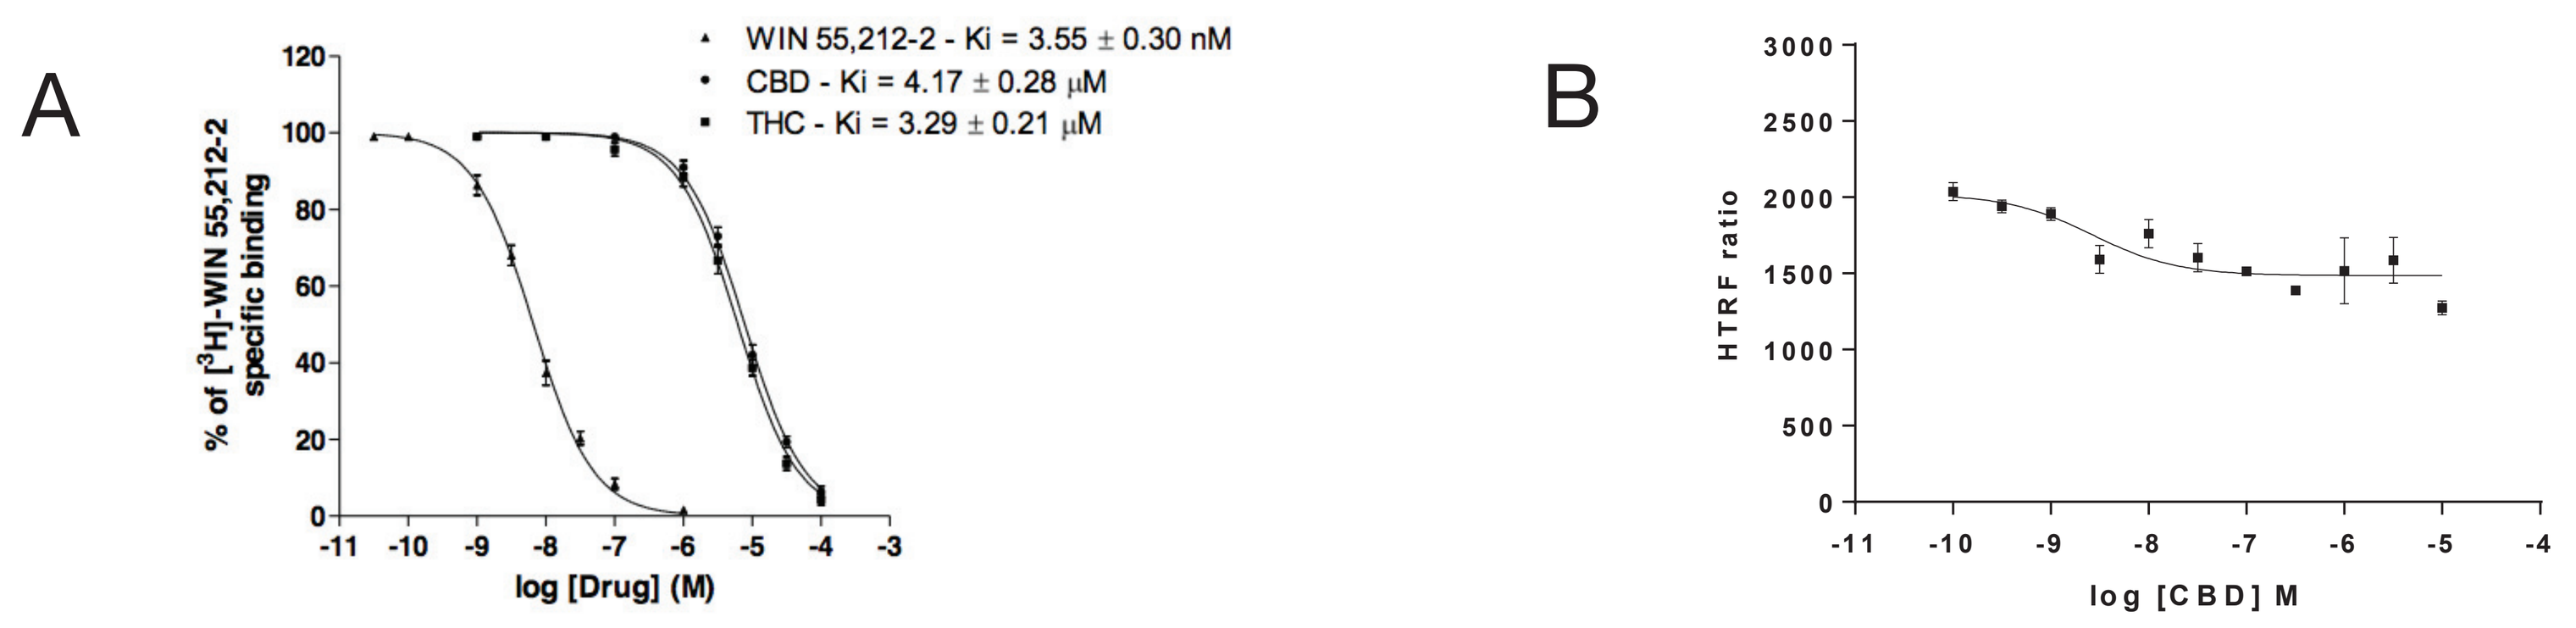
A
WIN 55,212-2 - Ki = 3.55 ± 0.30 nM
CBD - Ki = 4.17 ± 0.28 µM
THC - Ki = 3.29 ± 0.21 µM
% of [3H]-WIN 55,212-2 specific binding
log [Drug] (M)
B
HTRF ratio
log [CBD] M

Figure 2. Saturation and association/dissociation curves measured by HTRF
(A) Saturation binding experiments of red CB_2R ligand (CM-157) in HEK-293T cells transiently transfected with SNAP-CB_2R in the presence of 0 nM (circles), 10 nM (squares), or 100 nM (triangles) CBD. (B-C) Association/dissociation curves of 24 and 48 nM red CB_2R ligand, respectively, in the presence and absence of 5 µM CBD. Data represents the mean ± SEM of a representative experiment performed in quadruplicate. HTRF ratio = 665 nm acceptor signal/620 nm donor signal x 10,000. Kinetic constants and dissociation half-time ($t_{1/2}$) are indicated in table 1

Table 1 Kinetic parameters from dissociation experiments performed with two different concentrations of red CB2R ligand calculated from HTRF data.

	24 nM red CB_2R ligand		48 nM red CB_2R ligand	
Compound	k_{off} (s^{-1})	$t_{1/2}$ (s)	k_{off} (s^{-1})	$t_{1/2}$ (s)
CBD	0.0028	244	0.0028	251
Unlabeled CB_2R ligand	0.0055	126	0.0055	126
Unlabeled CB_2R ligand + CBD	0.0047	148	0.0045	153

k_{off}, dissociation constant
$t_{1/2}$, half life

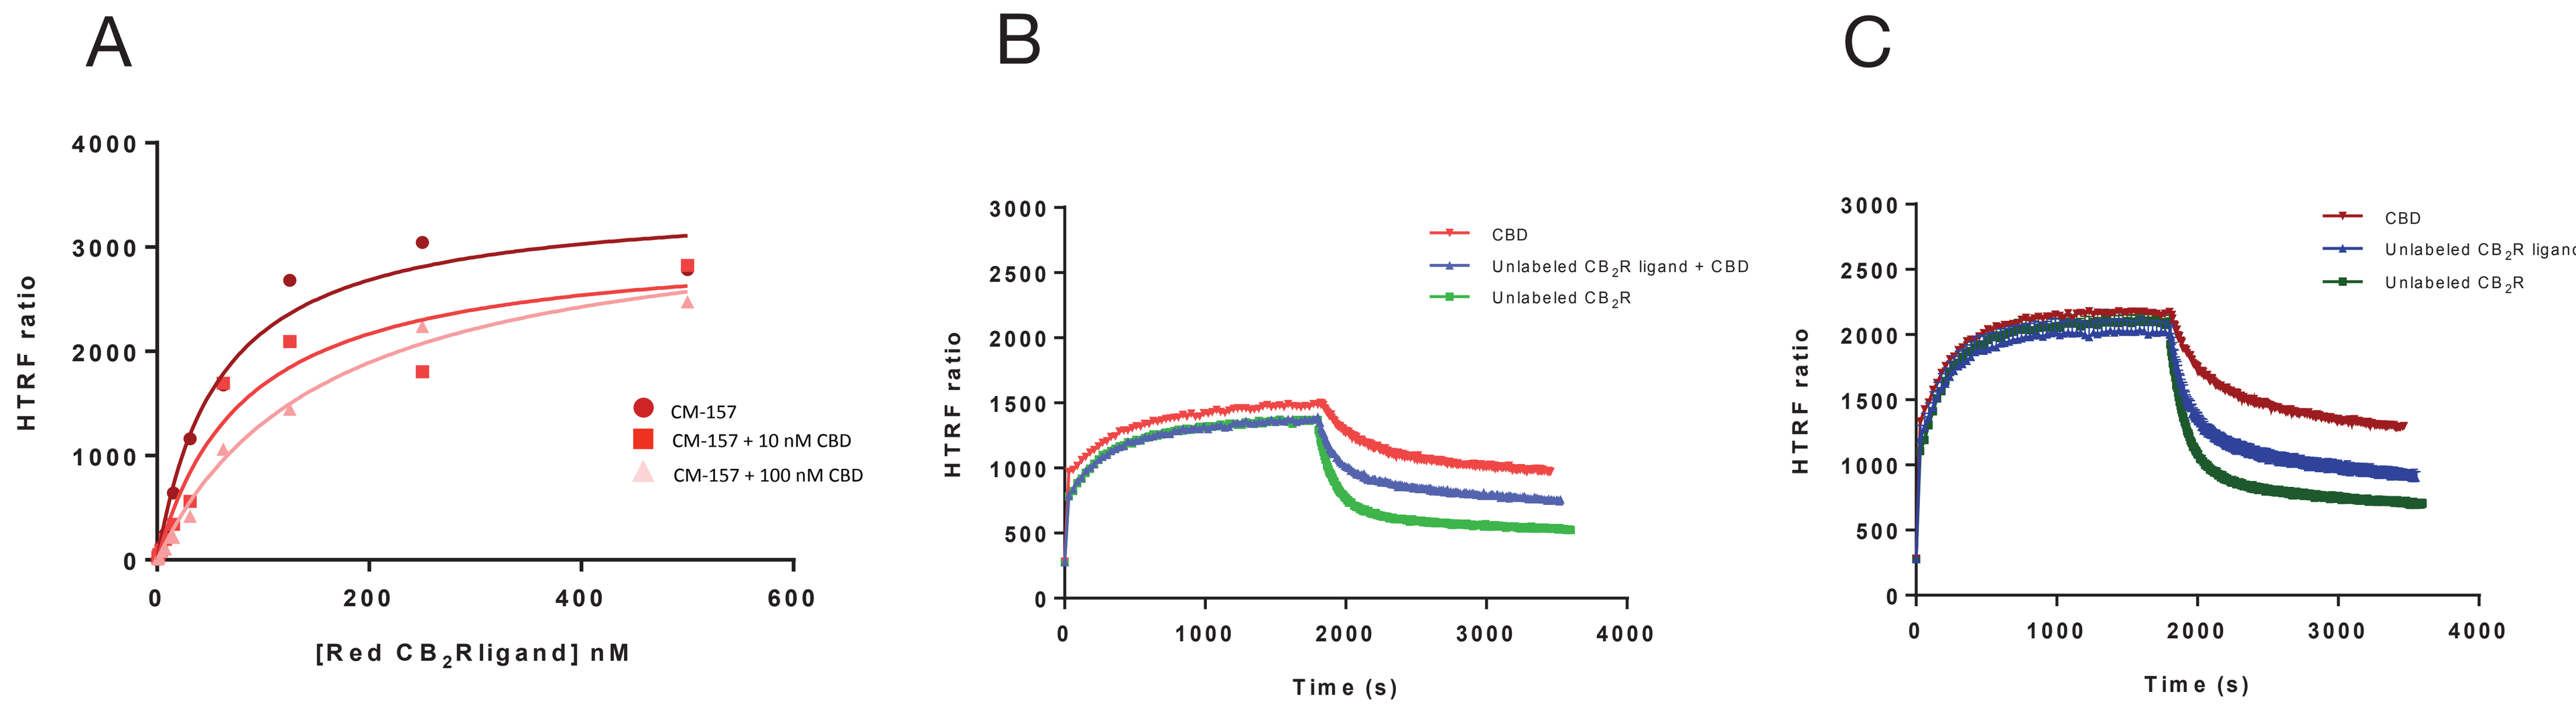
A
HTRF ratio
CM-157
CM-157 + 10 nM CBD
CM-157 + 100 nM CBD
[Red CB2R ligand] nM
B
CBD
Unlabeled CB2R ligand + CBD
Unlabeled CB2R
Time (s)
C
CBD
Unlabeled CB2R ligand + CBD
Unlabeled CB2R
Time (s)

Figure 3. cAMP assays
HEK-293T cells were transiently transfected with cDNA encoding for human CB_2 (A) or GPR55 (B) receptors. The effect of CBD on the decrease by JWH133 of forskolin-induced cAMP levels is displayed as a dose-response curve (panel A, right). Cells were pretreated 0.5 µM forskolin (15 minutes) and subsequently treated (15 minutes) with different concentrations of CBD in the absence or presence of 100 nM JWH133, a selective CB_2R agonist. Data (mean ± SEM) are given in percentage of the 500 nM forskolin-induced cAMP concentration. Significant differences were analyzed on data from seven different experiments; one-way ANOVA and Bonferroni's multiple comparison post hoc test were used for statistical analysis ($F_{4,71}$= 15, $p<0.001$) (**p<0.01, ***p<0.001, versus 0 nM CBD treatment).

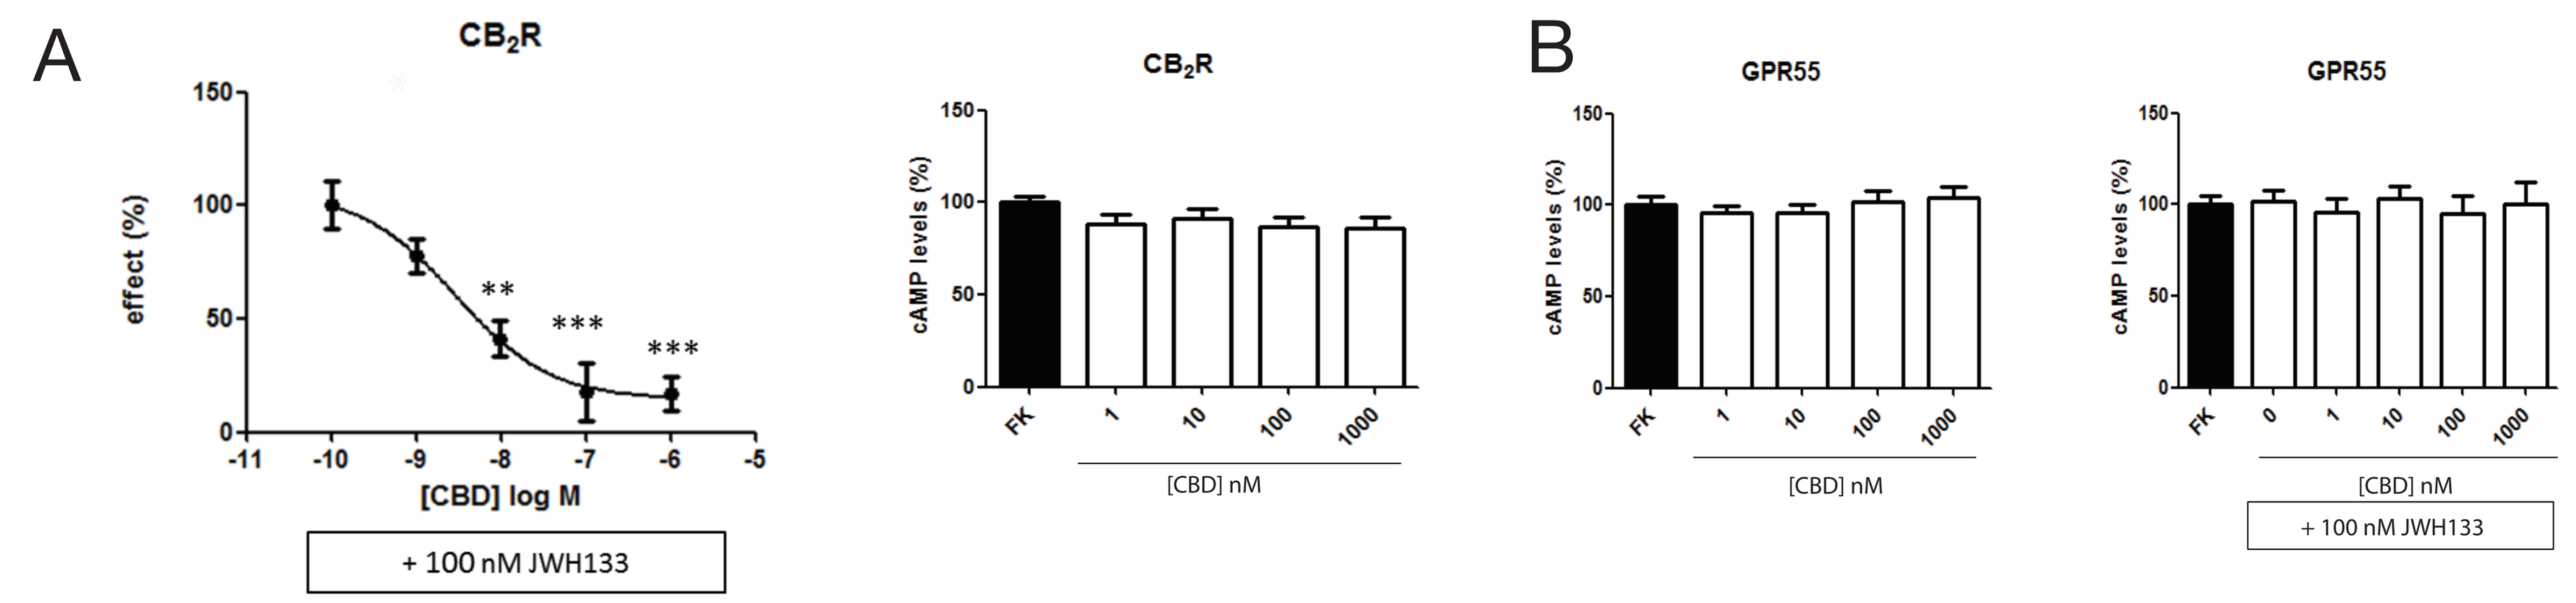
A
CB2R
effect (%)
[CBD] log M
+ 100 nM JWH133
CB2R
cAMP levels (%)
[CBD] nM
B
GPR55
cAMP levels (%)
[CBD] nM
GPR55
[CBD] nM
+ 100 nM JWH133

Figure 4. Hypothesis of action of CBD at allosteric site of CB_2R

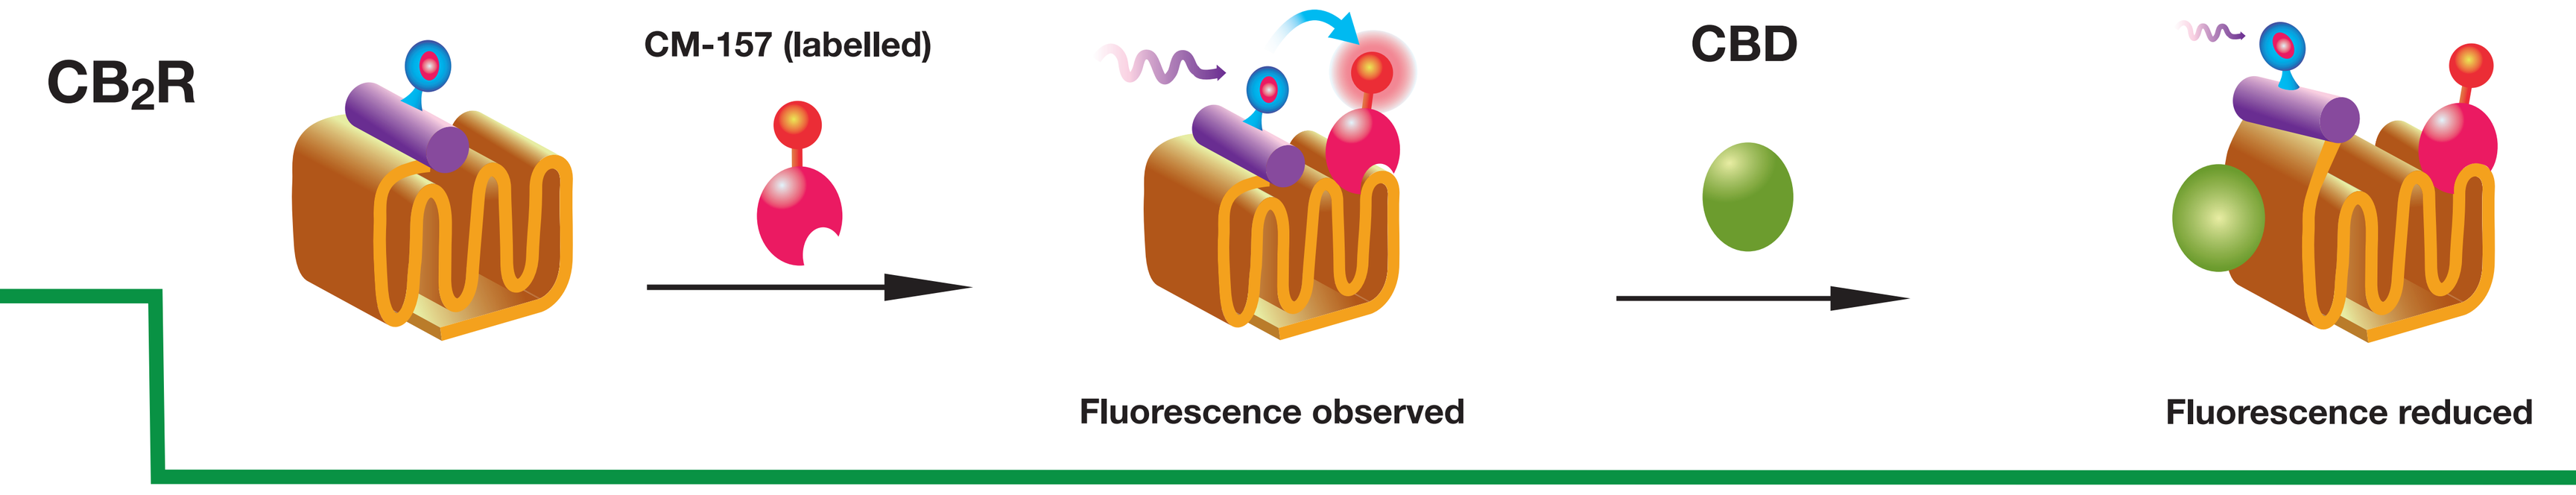
CB2R
CM-157 (labelled)
CBD
Fluorescence observed
Fluorescence reduced

CONCLUSION AND DISCUSSION

The results here reported confirm that CBD is not able to displace the binding of WIN 55,212-2 to the orthosteric centre of the CB_2R. The HTRF data, however, showed that the binding of the labelled CM-157 to HEK-293T cells expressing CB_2R was slightly but consistently modified by nanomolar concentrations of CBD. Moreover, an effect of CBD on dissociation kinetics was not appreciated in radioligand but in HTRF binding studies. The most plausible interpretation of these data is an allosteric effect disclosed in HTRF-mediated binding assays and a slightly different binding mode of radiolabeled WIN 55,212-2 or fluorescence-labelled CM-157 to CB_2R. This hypothesis was confirmed by functional experiments in which the effect of a selective CB_2R agonist was modulated by CBD at physiologically relevant concentrations.

Our results are in concordance of previous findings that CBD acts as an antagonist at the CB_1R and CB_2R at nanomolar range in vitro (Thomas et al., 2007). In addition we show for the first time with the present results that CBD at nanomolar range is able to modify the binding (Figure 1B), reduce the saturation of binding (Figure 2A) of the fluorescent labeled CM-157 at CB_2R, and inhibit the response of inhibition of forskolin-induced cAMP produced by the selective CB_2R agonist JWH-133 (Figure 3A). Accordingly, CBD was suspected as acting as allosteric modulator of cannabinoid receptors (Rimoldi and Bow, 2016; Laprairie et al., 2015).

Allosteric modulators of natural origin do usually provide negative modulation in both enzymology and pharmacology. It is noteworthy that CBD treatment leads to negative actions while the half-life of the receptor-agonist complex is longer in the presence of CBD (table 1). Moreover, finding allosteric CBD binding motives the two cannabinoid receptors would help in understanding some of the actions reported for CBD.

ACKNOWLEDGMENTS

This research was supported by the SAF2012/39875-C02-01 grant from the Spanish Ministry of Economy and Innovation, and by grants 201413330 and 201441331 from Fundació La Marató de TV3.

REFERENCES

Laprairie *et al.*, Br. J. Pharmacol. 172, 4790–4805 (2015).
Rimoldi *et al.*, Perspect. Medicin. Chem. 8, 17 (2016).
Thomas *et al.*, Br. J. Pharmacol. 150, 613–623 (2007).

Phytoplant Research
www.phytoplant.es